3.269

ALTERNATE DOSING REGIMENS FOR AN IMMEDIATE RELEASE FORMULATION OF THE NOVEL ANTISEIZURE MEDICATION LP352 BASED ON PHARMACOKINETICS (PLASMA AND CEREBROSPINAL FLUID [CSF]) AND PHARMACODYNAMICS (QEEG)

Randall Kaye,[1] Nuggehally Srinivas,[1] Dewey McLin,[1] Chad Orevillo,[1] David Vossler,[2] Rosa Chan[1]

[1]Longboard Pharmaceuticals, La Jolla, CA, USA
[2]University of Washington, Seattle, WA, USA

©Longboard Pharmaceuticals.
All Rights Reserved.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission of the authors.

Scan to download a reprint of this poster.

Acknowledgments

This study was sponsored by Longboard Pharmaceuticals, Inc. (La Jolla, CA, USA). Medical writing assistance was provided by ApotheCom (San Diego, CA, USA) and funded by Longboard Pharmaceuticals.

Presented at the Annual Meeting of the American Epilepsy Society; December 1-5, 2023; Orlando, FL

BACKGROUND

- LP352 is a potent and highly selective 5-hydroxytryptamine (5-HT_{2C}) superagonist designed for the treatment of individuals with developmental and epileptic encephalopathies (DEEs)
 - LP352 elicits a greater response than serotonin at the 5-HT_{2C} receptor and has no detected activity at receptors associated with significant adverse side effects: 5-HT_{2A} (psychiatric: insomnia, hallucinations, euphoria)[1] and 5-HT_{2B} (valvular heart disease and pulmonary arterial hypertension)[2,3]
- Relative to dosing 3 times a day (TID),[4] dosing twice daily (BID) in patients with DEEs may lower the therapeutic burden by increasing patient adherence to treatment and easing delivery of treatment for caregivers

OBJECTIVE

- To evaluate the pharmacokinetics (PK) and pharmacodynamics (PD) of TID and BID doses of LP352

METHODS

- This was a phase 1, open-label, multiple-dose study in healthy adults between 18 and 55 years of age (n = 25; **Table 1**)
- Following an initial 2 to 4 days of up-titration (depending on target dose), volunteers were dosed for 6 to 8 days at the target doses of 12 mg, 15 mg, or 18 mg BID
- Serial plasma samples were obtained after a single dose and after multiple doses on days 8, 9, and 10
- Serial cerebrospinal fluid (CSF) samples were obtained at steady state on day 11
- Serial quantitative electroencephalograms (QEEGs) with eyes in the open and closed positions were recorded at baseline and days -1, 1, 3 or 5, 10, and 16 (at trough)
- LP352 concentrations were quantified in various clinical samples using validated bioanalytical methods
- Standard PK parameters were calculated
- All QEEG assessments used state-of-the-art recording instruments and were performed by qualified electroencephalogram technologists
- 18 mg BID exposure data was compared to previously reported 12 mg TID exposure data[4]

Table 1. Schedule of Dosing

- Serial EEGs
- Serial PK samples
- Serial CSF samples

Up-titration phase | Target dosing | Taper-down phase

Cohort (n = 25)	Dose #	D-1	D1	D2	D3[b]	D4	D5[b]	D6	D7	D8	D9	D10	D11	D12	D13	D14	D15	D16
3 (12 mg BID)	1	–	6	6	12	12	12	12	12	12	12	12	12	6	3	3	3	3
	2	–	6	6	12	12	12	12	12	12	12	12	6	6	3	3	–	–
4A (15 mg BID)[a]	1	–	6	6	12	12	15	15	15	15	15	15	15	9	4.5	4.5	4.5	4.5
	2	–	6	6	12	12	15	15	15	15	15	15	9	9	4.5	4.5	–	–
4B (18 mg BID)[a]	1	–	6	6	12	12	18	18	18	18	18	18	18	9	4.5	4.5	4.5	4.5
	2	–	6	6	12	12	18	18	18	18	18	18	9	9	4.5	4.5	–	–

[a]A minimum of 4 subjects (Cohort 4a) were up-titrated to 15 mg BID dose on day 5 and completed the day 11 morning dose before the remaining subjects (Cohort 4b) were up-titrated to the target 18 mg BID dose.
[b]Serial QEEGs were obtained on days 3 or 5.

RESULTS

Participants

- 25 healthy adult volunteers were included in this study (20 male and 5 female volunteers)
- The mean weight of volunteers was 73.5 kg, 77.8 kg, and 75.2 kg, for LP352 12 mg, 15 mg, and 18 mg BID, respectively

Pharmacokinetics

- Across BID dose ranges, the observed PK in both plasma and CSF was dose linear (**Table 2**)
- The CSF/plasma exposure ratio for area under the concentration-time curve from time zero to the end of the dosing interval (AUC_{tau}) was generally >0.84 at the 3 BID dose levels (**Table 2**)

Table 2. Summary Statistics of LP352 12 mg, 15 mg, and 18 mg BID Plasma and CSF PK Parameters

Dose (mg)	AUC_{tau} in CSF (h*ng/mL)	AUC_{tau} in Plasma (h*ng/mL)	CSF/Plasma Ratio[a]
12 mg BID	165.1	169.7	0.973
15 mg BID	175.6	200.4	0.876
18 mg BID	210.1	235.6	0.892

[a]Computed using the CSF AUC_{tau}/plasma AUC_{tau} (total).

- There was a strong correlation of plasma and CSF PK parameters, including maximum concentration (C_{max}) and AUC_{tau} (**Figure 1**)

Figure 1. AUC_{tau} (A) and C_{max} (B) of LP352 12 mg, 15 mg, and 18 mg BID in Plasma PK Samples

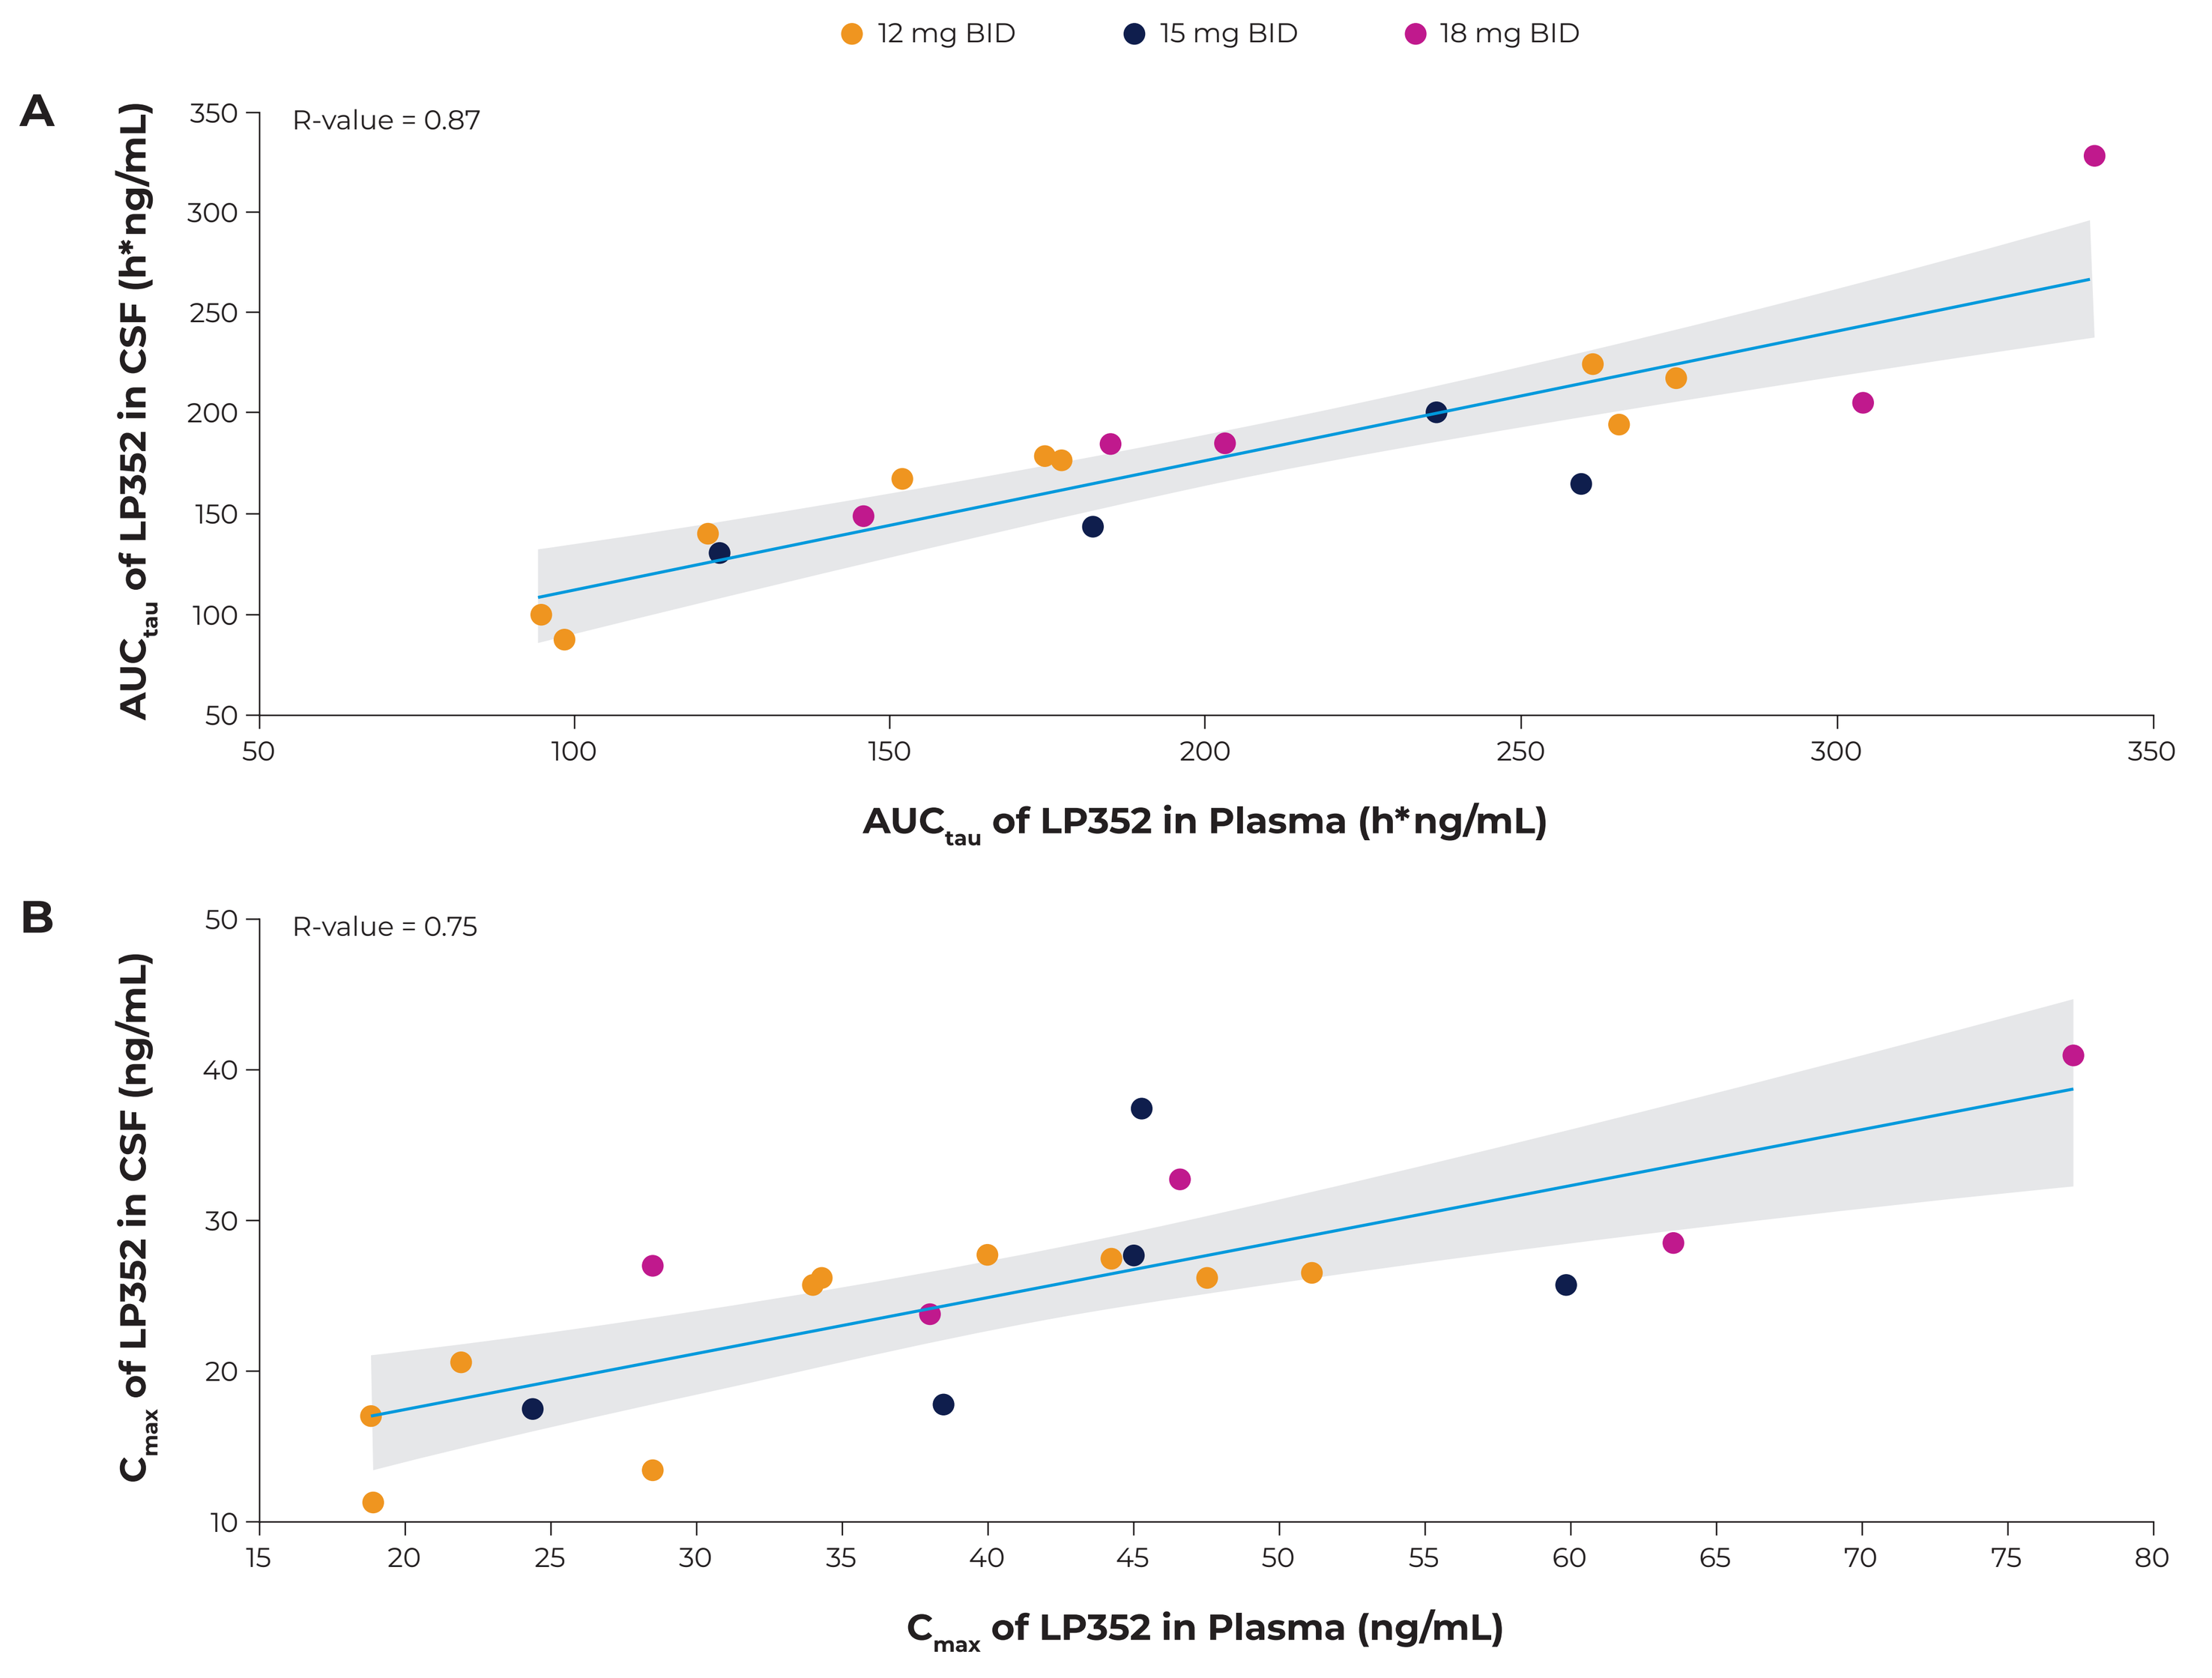
12 mg BID
15 mg BID
18 mg BID
A
R-value = 0.87
AUCtau of LP352 in CSF (h*ng/mL)
AUCtau of LP352 in Plasma (h*ng/mL)
B
R-value = 0.75
Cmax of LP352 in CSF (ng/mL)
Cmax of LP352 in Plasma (ng/mL)

- Average plasma concentrations over the dosing interval were comparable (~28%) between the 18 mg BID and 12 mg TID dose groups (**Figure 2**)

Figure 2. Plasma Average (A) and Maximum Concentrations (B) of LP352 12 mg TID and 18 mg BID

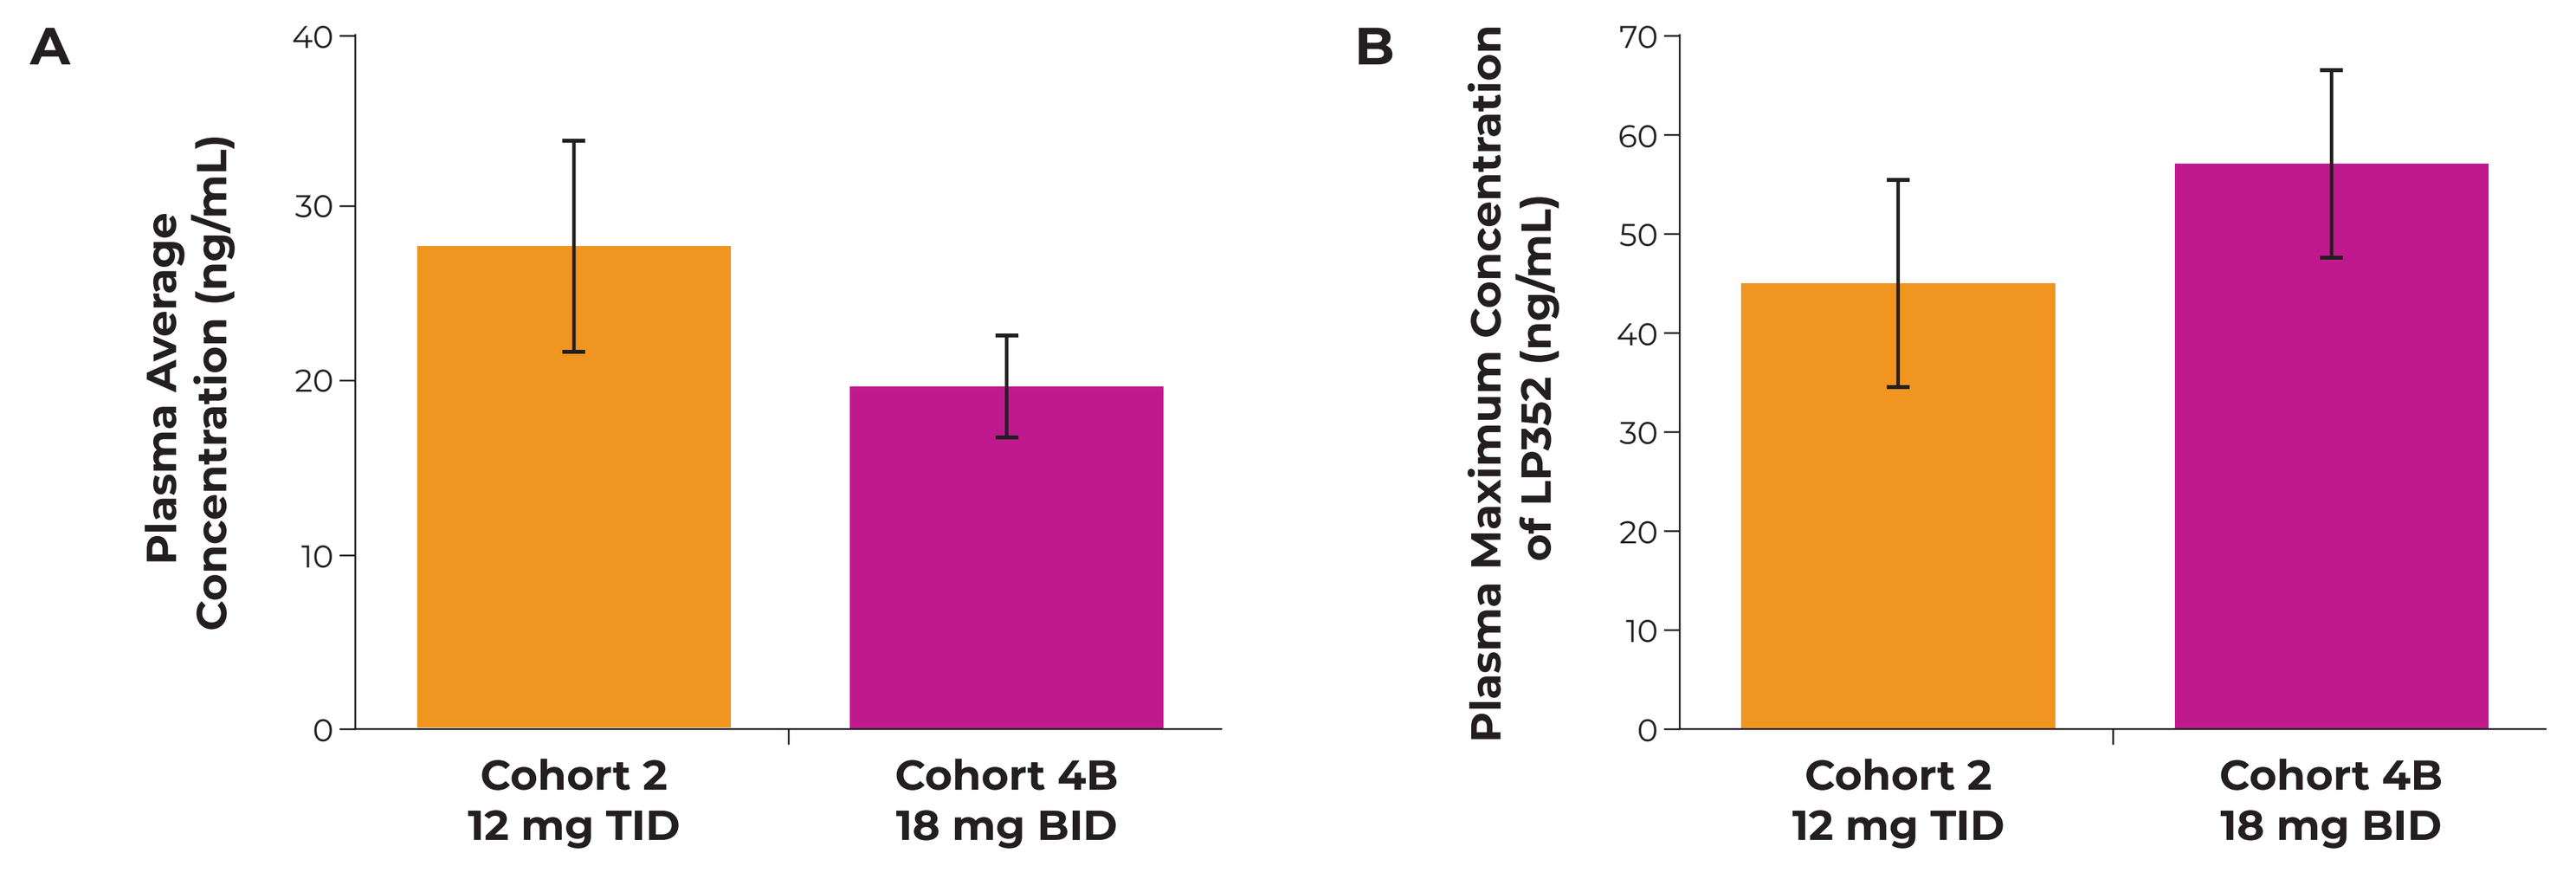
A
Plasma Average Concentration (ng/mL)
Cohort 2
12 mg TID
Cohort 4B
18 mg BID
B
Plasma Maximum Concentration of LP352 (ng/mL)
Cohort 2
12 mg TID
Cohort 4B
18 mg BID

- In the CSF, LP352 concentrations generally exceeded the Ki value (~14 ng/mL) for 5-HT_{2C} agonism by ~2-fold in both the 18 mg BID and 12 mg TID dosing groups (**Figure 3**)

Figure 3. CSF Average (A) and Maximum Concentration (B) of LP352 12 mg TID and 18 mg BID

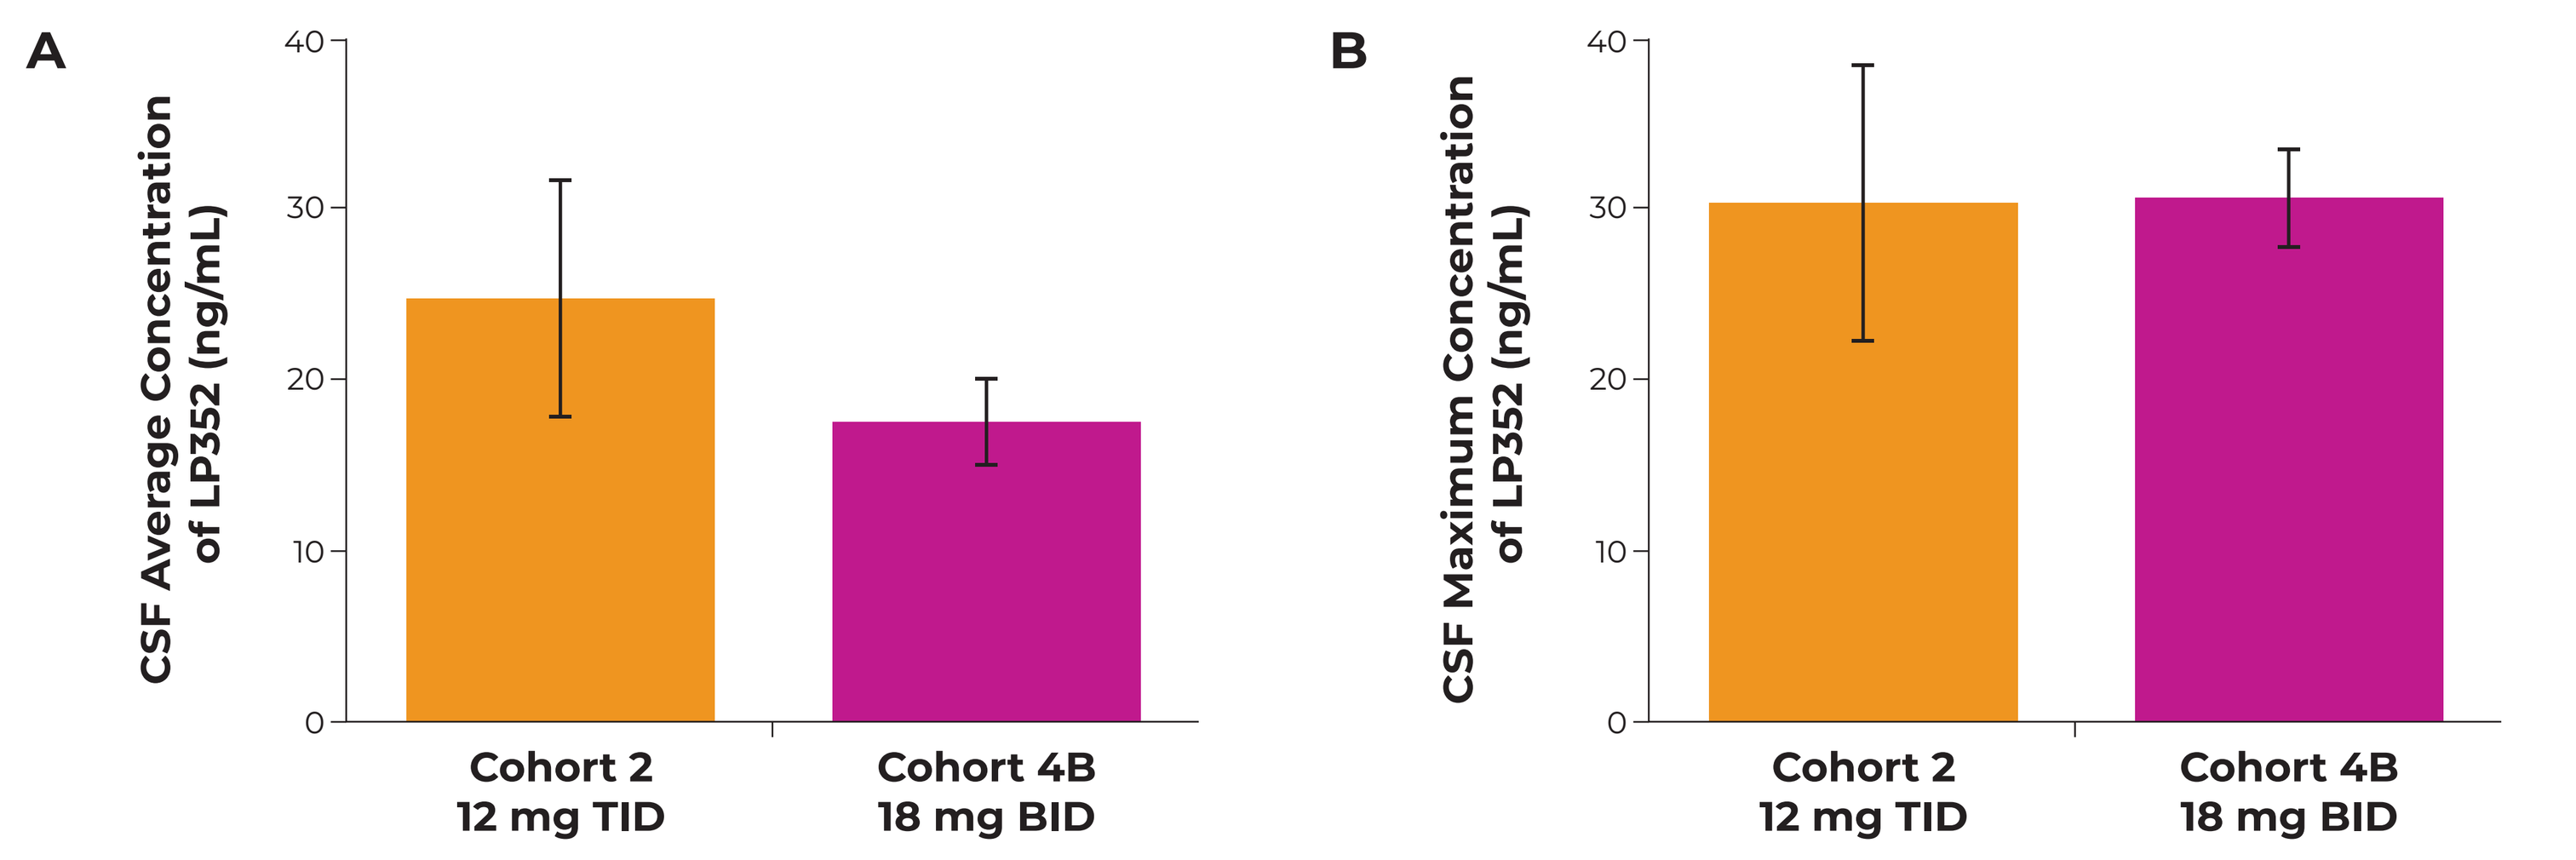
A
CSF Average Concentration of LP352 (ng/mL)
Cohort 2
12 mg TID
Cohort 4B
18 mg BID
B
CSF Maximum Concentration of LP352 (ng/mL)
Cohort 2
12 mg TID
Cohort 4B
18 mg BID

Pharmacodynamics

- Similar to TID dosing, the QEEG data across multiple spectral bands for BID dosing of LP352 was consistent, with successful receptor engagement in the brain,[4] and as observed previously, there was evidence of time and dose dependency of the effect

Safety

- The 3-step up-titration approach to reach the highest target dose of 18 mg BID was found to be safe and tolerable, with patients experiencing generally mild to moderate treatment-emergent adverse events (TEAEs)
- The most common TEAEs were nausea, vomiting, headache, and dizziness, which are consistent with earlier TID studies[4]
- There was a single serious adverse event, which was not drug-related
- Overall, the safety and tolerability of BID dosing of LP352 was comparable to TID dosing regimen as previously observed[4-6]

CONCLUSIONS

- **The plasma and CSF PK data of LP352, in conjunction with the observed PD QEEG changes, demonstrate that BID dosing of LP352 is a viable alternative option for use in future clinical studies**
- **Using an up-titration dose escalation scheme, the safety, tolerability, and general TEAE profile of LP352 for the BID regimen were comparable to those previously observed for the TID regimen**

Abbreviations **5-HT**, 5-hydroxytryptamine; **AUC_{tau}**, area under the concentration-time curve from time zero to the end of the dosing interval; **BID**, twice daily; **C_{max}**, maximum serum concentration; **CSF**, cerebrospinal fluid; **D**, day; **DEE**, developmental and epileptic encephalopathy; **PD**, pharmacodynamics; **PK**, pharmacokinetics; **QEEG**, quantitative electroencephalogram; **TEAE**, treatment-emergent adverse event; **TID**, 3 times daily.

References **1.** López-Giménez J, González-Maeso J. *Curr Top Behav Neurosci.* 2018;36:45-73. **2.** Higgins GA et al. *Pharmacol Ther.* 2020:107417. **3.** Hutcheson JD et al. *Pharmacol Ther.* 2011;132:146-157. **4.** Srinivas N et al. *Clin Pharmacol Drug Dev.* 2023;12(1):52-53. **5.** Parasrampuria D et al. *Neurology.* 2022;98(18):1750. **6.** Parasrampuria D et al. *Neurology.* 2022;98(18):1771.